第29屆台灣癌症聯合年會
The 29th Taiwan Joint Cancer Conference
Fueling Progress and Revolutionizing Cancer Care

【C6-004】

2025 TJCC

A Retrospective Study Comparing the Efficacy of Conventional Standard Care and ON101 in the Management of Radiation Dermatitis
比較傳統標準照護與ON101於放射線皮膚炎成效之回溯性研究

Hsiao-Lin Liu[1]; Yuk-Wah Tsang[1,2] (Correspondence); Sheng-You Su[3]; Hsiao-Ju Fu[1]; Chih-Chia Chang[1]

1. Dept. of Radiation Oncology Ditmanson Medical Foundation Chia-yi Christian Hospital
2. Dept. of Biomedical Engineering, Chung Yuan Christian University
3. Clinical Medicine Research Center, Ditmanson Medical Foundation Chia-yi Christian Hospital

Purpose:

Radiation dermatitis (RD) is a common acute side effect of radiation therapy, particularly when the target volume involves the skin or is near the skin surface, as seen in head and neck, breast, extremity sarcoma, and anal cancers. Conventional RD treatments for severe cases (Grades 3 and 4) include Silvadene ointment or Duoderm dressing to prevent infections and support natural wound healing. A newer treatment, ON101 cream, contains 1.25% extracts of *Plectranthus amboinicus* and *Centella asiatica*. Originally developed for diabetic ulcers, ON101 promotes wound healing by modulating macrophage polarization. It inhibits inflammation and shifts the wound microenvironment from M1 (pro-inflammatory) to M2 (anti-inflammatory) macrophage dominance. M2 macrophages enhance angiogenesis, stem cell recruitment, fibroblast proliferation, and collagen synthesis, thereby facilitating tissue regeneration and healing. We utilized ON101 off-label for RD management and observed promising results. This retrospective study compares the outcomes of conventional RD management with ON101 treatment.

Materials and Method:

Patients with RTOG Grade 3 or higher RD were included if they had complete treatment records. The study began when radiotherapy was suspended due to RD, with dressing applications continuing until the RTOG score improved to Grade 2 or lower. Patients who could not comply with the dressing regimen or had incomplete records were excluded. Medical records were analyzed to collect demographic and clinical data, including age, sex, BMI, cancer type and stage, surgical history, and maximum radiation dose. Treatment details, such as wound dressing type, RTOG grading, and VAS pain scores, were also recorded. The primary outcome measures included dressing duration, RTOG score improvement from Day 1 to Day 9, VAS improvement from Day 1 to Day 9, and overall VAS improvement from the first to the last dressing change. Statistical analyses, including Wilcoxon rank-sum tests, Fisher's exact tests, and Chi-square tests, were conducted to compare demographic and clinical characteristics between dressing groups. Multiple regression analysis was performed to identify factors influencing treatment outcomes. All analyses were conducted using R software (version 4.4.1).

Result:

A total of 58 patients were enrolled, with 26 receiving conventional dressings and 32 treated with ON101. Female patients and those with higher BMI were more likely to receive ON101. Baseline characteristics were comparable between groups, with no significant differences in initial RTOG ($p = 0.770$) or VAS scores ($p = 0.262$).Significant differences emerged in treatment outcomes. **The ON101 group had a shorter median dressing duration (12 vs. 15.5 days; p = 0.003) and greater clinical improvement**. **By Day 9, ON101 users showed a median RTOG improvement of 1 grade**, whereas the conventional group showed no change ($p < 0.001$). **Pain scores also improved more in the ON101 group, with median VAS reductions of 7 points by Day 9 and 9 points overall**, compared to 2 and 6 points, respectively, in the conventional group ($p < 0.001$).Multiple regression analyses revealed that **dressing duration was significantly reduced in the ON101 group** ($\beta = -7.8$, $p = 0.002$), and influenced by initial VAS score ($\beta = 3.38$, $p = 0.001$) and BMI ($\beta = -0.62$, $p = 0.025$). RTOG improvement was positively associated with ON101 use ($\beta = -0.9$, $p < 0.001$), initial RTOG score ($\beta = -0.82$), initial VAS ($\beta = 0.38$), and BMI ($\beta = -0.06$). **VAS improvements were significantly influenced by ON101 use and BMI, with ON101 providing an additional 4.35-point (Day 1–9) and 2.42-point (final) reduction in pain**. Higher initial VAS also predicted greater pain relief.

Case illustration:

A 70 y/o woman has malignant sarcoma, received operation with +margin. She underwent post-operative RT up to 70Gy/35 fractions with grade 3 radiation dermatitis. She utilized ON101 and achieved rapid dermatitis resolution.

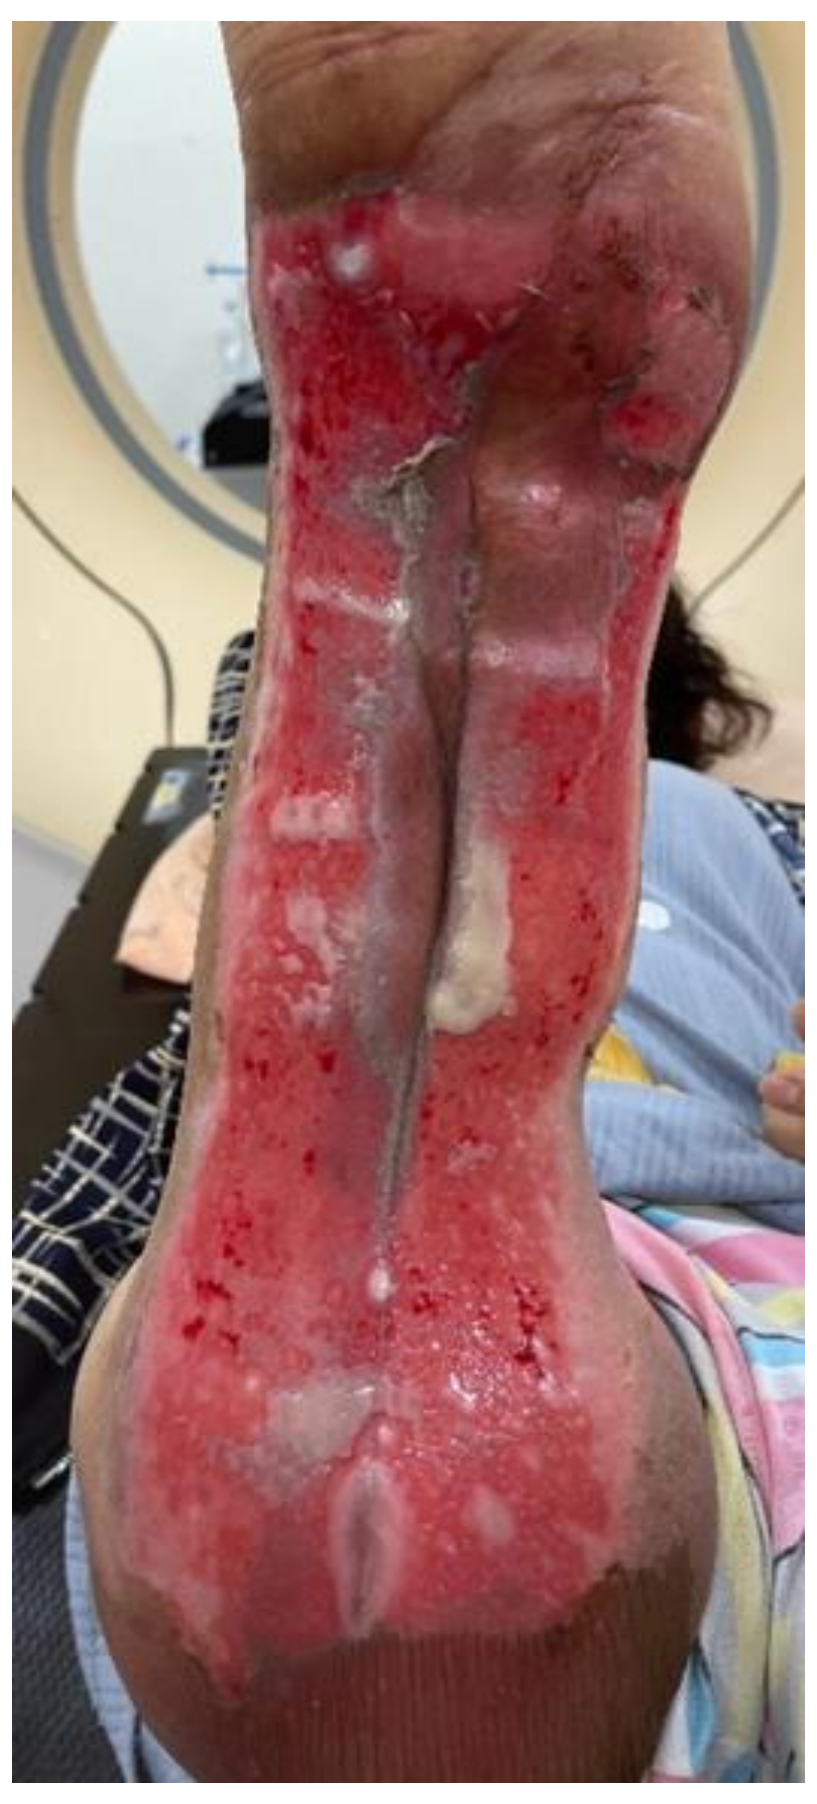

Day 1

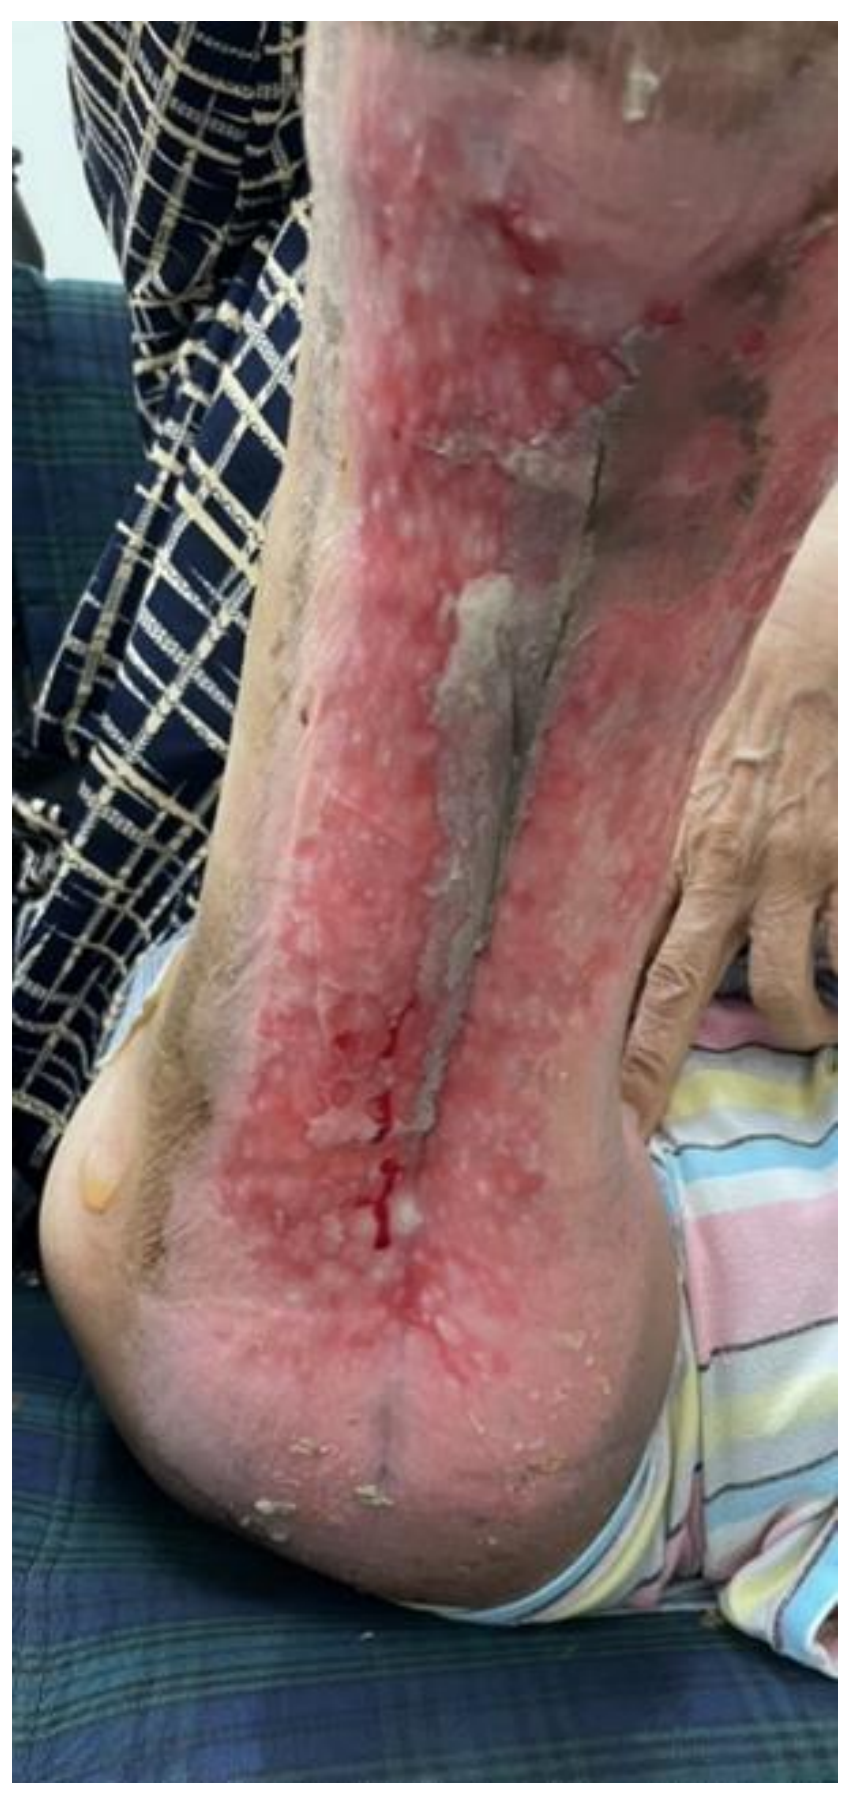

Day 4

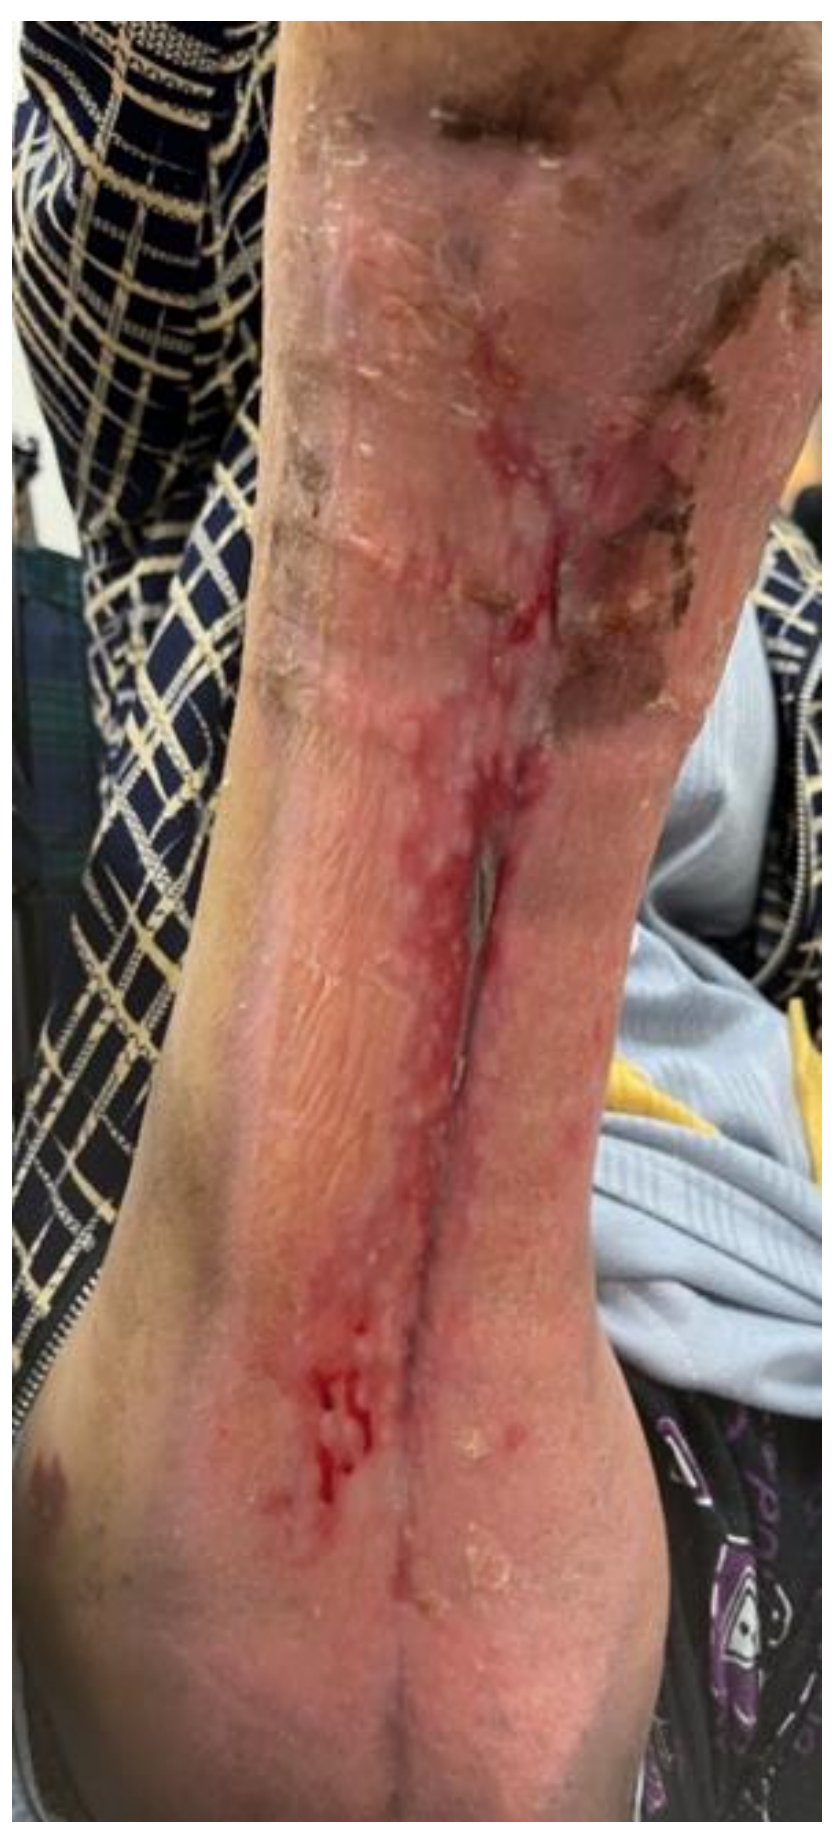

Day 11

Conclusion

ON101 dressing demonstrated superior performance compared to conventional dressings, significantly reducing dressing duration, enhancing wound healing, and alleviating pain. These findings suggest that ON101 may be an effective alternative for managing RD. A phase III randomized controlled trial should be conducted to confirm the efficacy of ON101 in RD management.