戴德森醫療財團法人
嘉義基督教醫院
CHIA-YI CHRISTIAN HOSPITAL

A CASE SERIES OF ON101 IN ACUTE RADIATION DERMATITIS

Hsiao-Ling Liu, RN (1st Author); Yuk-Wah Tsang, MD (Correspondence); Chih-Chia Chang, MD; Cheng-Yen Lee, MD; Yu-Wen Wang, MD
Dept. of Radiation Oncology, Chia-Yi Christian Hospital, Taiwan.

Introduction

Acute radiation dermatitis (ARD) is a common adverse effect of radiotherapy, particularly in cases where the skin is included in the target volume, such as head and neck, anal , extremity sarcoma or breast cancer. ARD severity ranges from mild erythema to moist desquamation and, in severe cases, necrosis. Current management strategies for mild to moderate ARD typically include interventions such as moist desquamation care, topical corticosteroids, and antibiotics. ON101, a novel topical macrophage regulator, has shown the ability to inhibit the NLRP3 inflammasome cascade and suppress pro-inflammatory cytokines through M1 macrophage modulation in preclinical studies. Thiscase series aimed to assess the clinical efficacy of ON101 in managing ARD.

Methods

Seven patients with grade 3 to grade 4 ARD were treated with ON101 cream, with secondary dressings applied as needed. The primary endpoints were the resolution of erythema and wound healing. Patient-reported outcomes, including treatment satisfaction and quality-of-life improvements, were also documented.

Results

Treatment with ON101 led to complete resolution of erythema in all patients (100%), with wound healing achieved within 4 to 15 days. Furthermore, seven patients reported improvements in quality of life following the intervention.

A 57 y/o lady with left breast cancer,
received 50Gy/25 fractions with grade 4 ARD.
She utilized ON101 3 grams and ARD down-graded to grade 1 in 8 days.

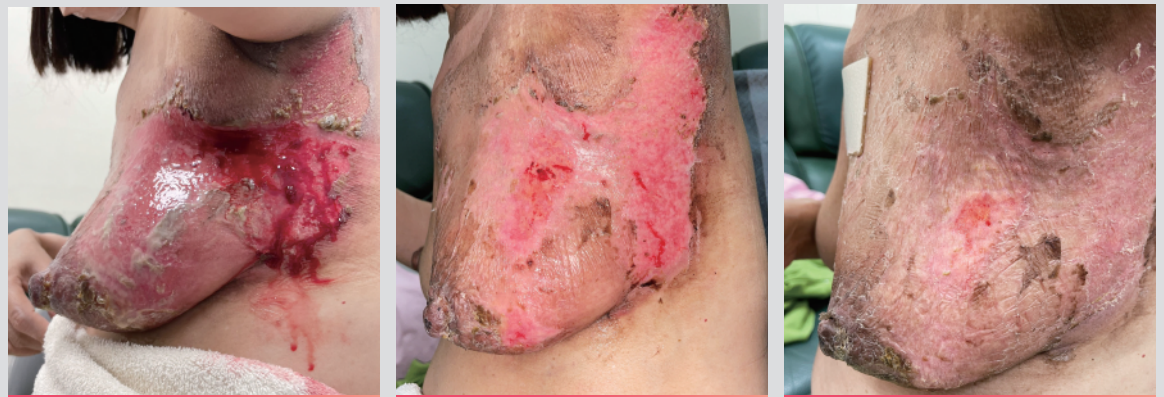

Day 1 | Day 5 | Day 8

A 63 y/o man had hypopharynxcancer,
received 70Gy/35 fractions irradiation with grade 4 ARD.
He utilized ON101 4.5 grams and ARD down-graded to grade 2 in 8 days.

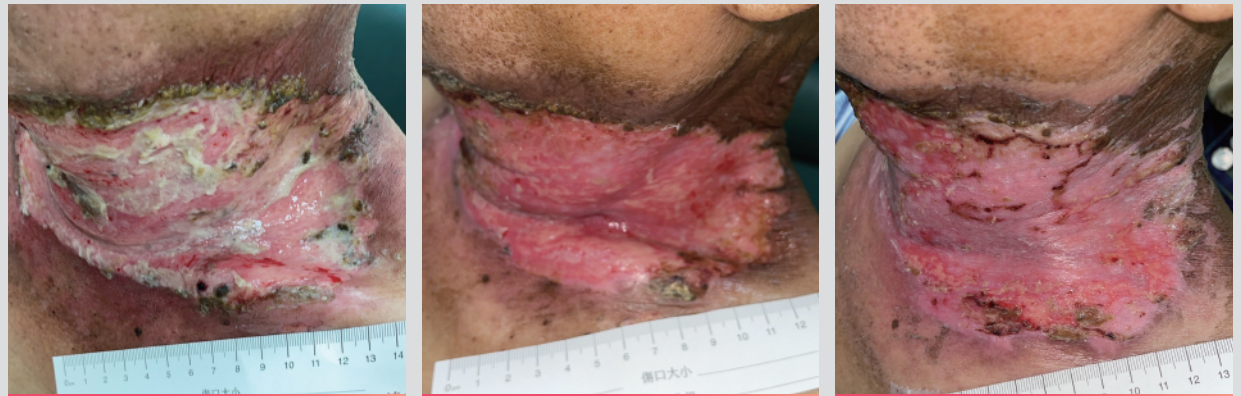

Day 1 | Day 5 | Day 8

Discussion

This case series highlights the successful translation of ON101's preclinical efficacy into clinical practice. By targetingthe NLRP3 inflammasome signaling pathway, ON101 effectively alleviated radiation-induced skin damage. These preliminary findings suggest that ON101 is a promising treatment option for ARD, providing rapid wound healing and enhanced patient outcomes. Further clinical trials are warranted to validate these results and explore its broader clinical application.

Presented at The Symposium on Advanced Wound Care (SAWC) Spring | Wound Healing Society (WHS), Grapevine, TX, Apr 30 - May 3, 2025